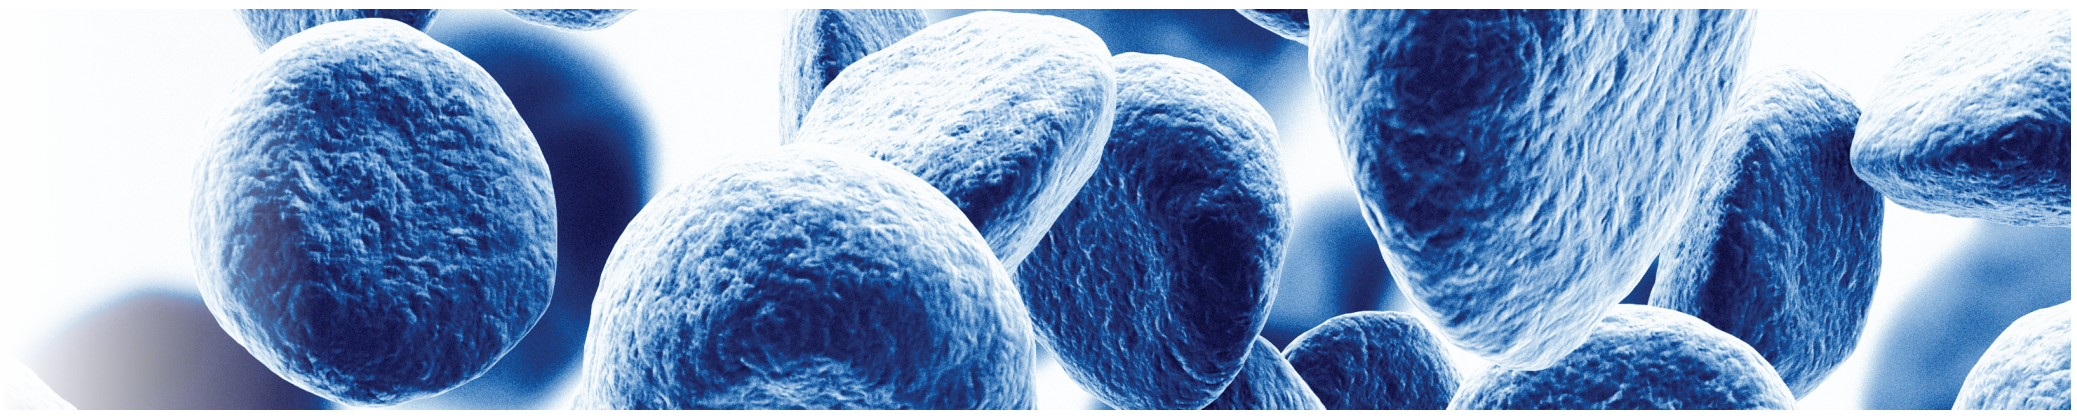

Abstract EBMT Meeting 2016

Leukemia-associated antigen reactive T-cells in ATIR101, a recipient-specific allodepleted T-cell product facilitating haploidentical HSCT

Jurjen Velthuis* [1], Richard Klar[2], Halvard Bönig[3], Jeroen Rovers[1], Denis-Claude Roy[4], Lea Prokop[5], Stefan Stevanović[5], Manfred Rüdiger[1], Angela M. Krackhardt[2]

[1] Kiadis Pharma, Amsterdam-Duivendrecht, Netherlands, [2] Medizinische Klinik III, Klinikum Rechts der Isar, TU Munich, Munich, [3] German Red Cross Blood Service Baden-Württemberg-Hesse, Frankfurt, Germany, [4] Hôpital Maisonneuve Rosemont, Montreal, Canada, [5] Institut für Zellbiologie, Eberhard Karls Universität, Tübingen, Germany

Introduction: Graft-versus-leukemia (GvL) relies on donor T-cells killing host leukemia cells post-transplant. T-cells with preferential recognition of leukemia cells have been well documented but their clinical relevance remains challenged because thymic selection prevents a high-affinity interaction between T-cells and antigen presenting cells (APC) expressing regular antigens in self-HLA. ATIR101, a personalized T-cell immunotherapeutic selectively depleted of HLA-haplotype mismatched T-cells, provides a unique platform to study leukemia-reactive T-cells as high-affinity interactions with antigens expressed in the mismatched haplotype may occur, whereas T-cells responding to the antigen-presenting foreign HLA-molecule have been eliminated.

Material (or patients) and methods: Two out of the first 10 ATIR101 batches manufactured in clinical phase 2 study CRAIR- 007 (NCT01794299) met the requirement of having a mismatched haplotype known the be able to express a known leukemia-associated antigen; these batches were used to screen for the presence of leukemia-associated antigen reactive T-cells. Because the frequency of leukemia-associated antigen reactive T-cells is expected to be very low, we used peptide-MHC monomers of the mismatched HLA-haplotype presenting leukemia-associated antigens and established a stimulation platform with artificial APCs (aAPC). Those aAPC consisted of streptavidin coated microspheres loaded with a biotinylated anti-CD28 antibody and the respective biotinylated peptide HLA monomer.

Results: In one of the two batches, leukemia-associated antigens specific T-cells were detected: ATIR101 cells were stimulated with Myb628 /HLA-B44 aAPCs and Myb628 specific T-cell expansion was assessed after one or two rounds of stimulation; an irrelevant HLA-B44 multimer was used as negative control. Clearly, we were able to detect CD8+ T-cells with specific reactivity against one HLA-B44- restricted leukemia-associated HLA ligand derived from the MYB gene (figure 1).

Conclusion: These data show that T-cells recognizing leukemia-associated antigens expressed in the mismatched HLA haplotype are retained in ATIR101 from which the T-cells responding to the antigen-presenting foreign HLA-molecule have been eliminated. Conceivably, these cells may contribute to the Graft-versus-leukemia (GvL) effect of ATIR101.

Abstract EBMT Meeting 2016

Figure 1:

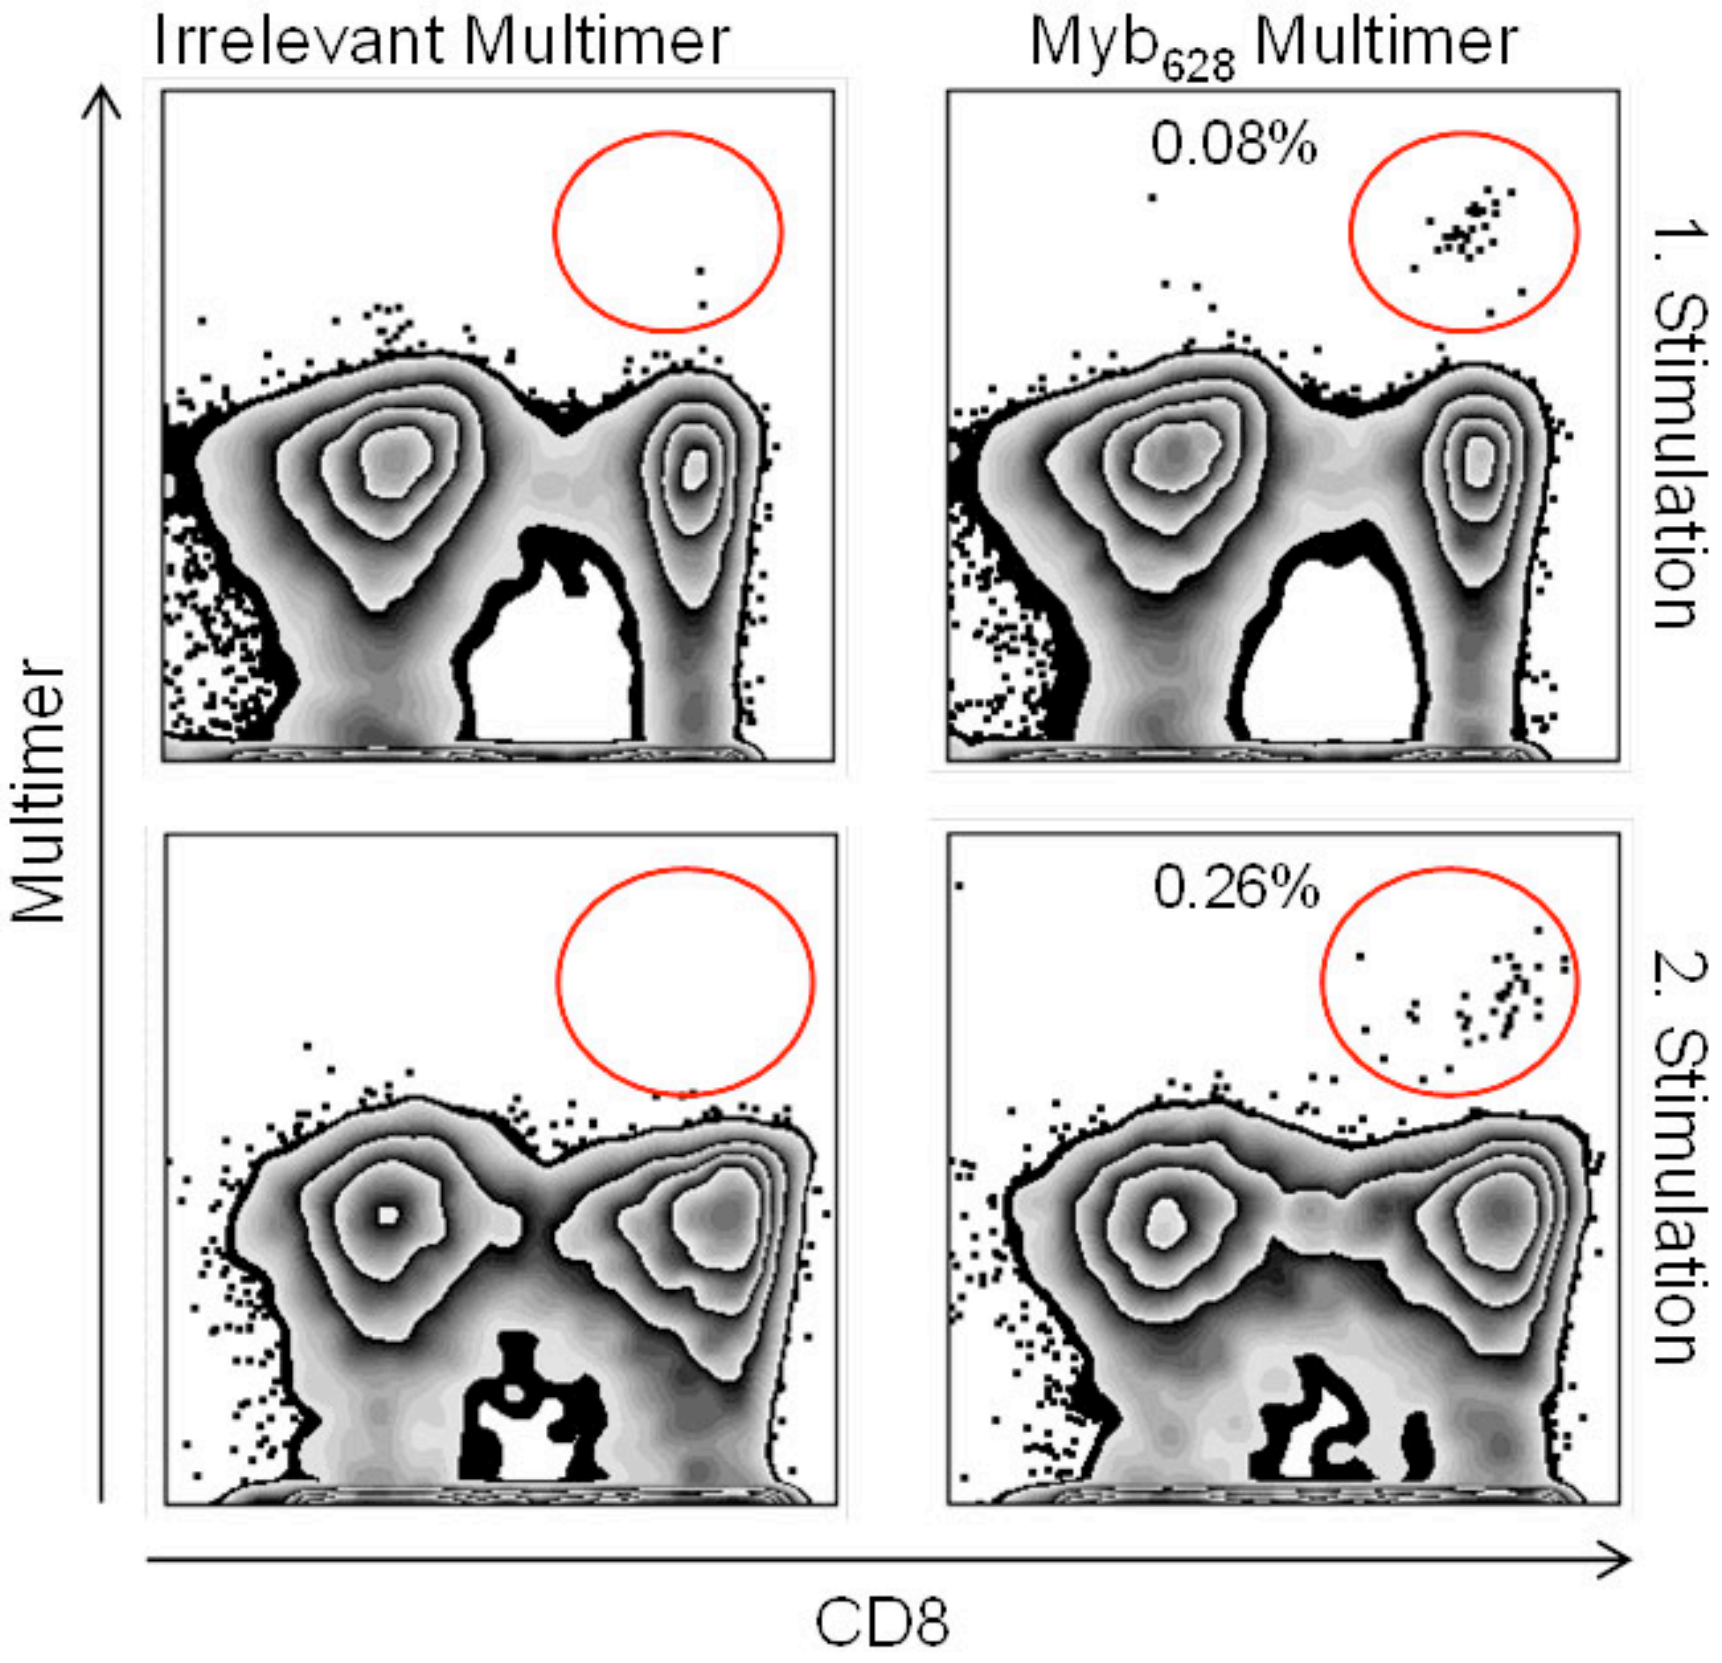
Irrelevant Multimer
Myb_{628} Multimer
0.08%
0.26%
1. Stimulation
2. Stimulation
Multimer
CD8